Comprehensive genomic profiling reveals novel opportunities for treatment-refractory gastrointestinal cancers

Wei Yen Chan[1], Subotheni Thavaneswaran[1,2,3], Frank Lin[2,3], Lucille Sebastian[3], Mandy Ballinger[2], John Grady[2], Maya Kansara[2], Anthony Joshua[1,2], Rasha Cosman[1], John Simes[3], David Thomas[1,2]

1 The Kinghorn Cancer Centre St Vincent's Hospital Sydney, 2 Garvan Institute of Medical Research, 3 NHMRC Clinical Trials Centre

Background

- Comprehensive genomic profiling (CGP) can reveal targetable oncogenic drivers to inform treatment options beyond traditional chemotherapeutics based purely on tumour histology and origin.
- This is a case series from the Cancer Molecular Screening and Therapeutics (MoST) program, which employs molecular screening to identify potential actionable mutations and corresponding treatment[1].

Objectives

- To illustrate how genomic findings can offer novel therapeutic opportunities and improve outcomes for patients with treatment-refractory gastrointestinal (GI) cancer.

Methods

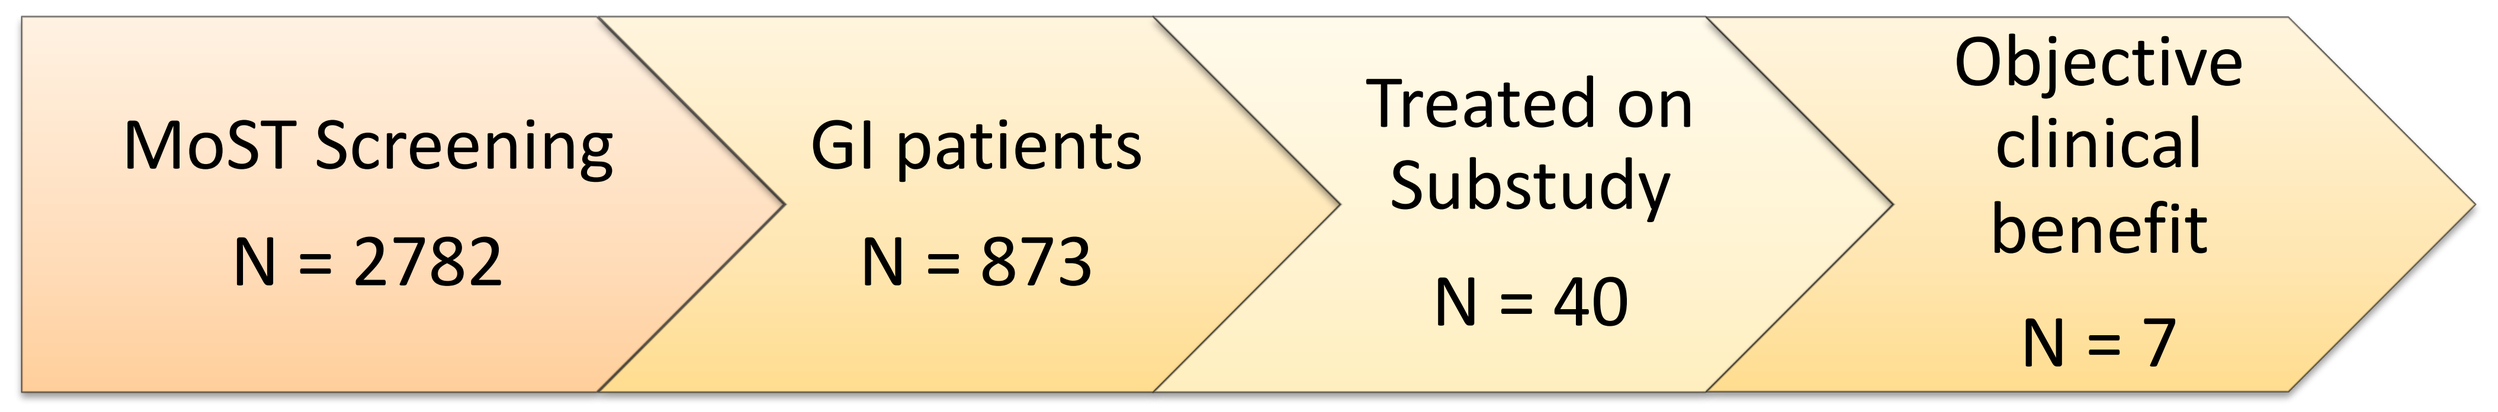
MoST Screening
N = 2782
GI patients
N = 873
Treated on Substudy
N = 40
Objective clinical benefit
N = 7

- Patients with advanced solid tumours of any histology undergo molecular screening.
- Genomic alterations are reviewed at molecular tumour boards to identify appropriate biomarker-matched therapeutic substudies.
- Seven GI cancer patients with durable clinical benefit were identified by case review.

Results

Case 1

- *POLE* encodes DNA polymerase ε, responsible for leading strand DNA replication[2]. *POLE* mutations result in accumulation of DNA errors, genetic instability and high TMB.
- A patient with pancreatic acinar cell carcinoma (1-2% of pancreatic neoplasms) was treated with durvalumab + tremelimumab following identification of underlying *POLE P286R* mutation and high TMB (204 mutations/megabase).
- After 7 cycles of study treatment, there was a near complete radiological response in the liver metastasis (Figures 1 and 2), permitting liver metastasectomy, which revealed a complete pathological response.

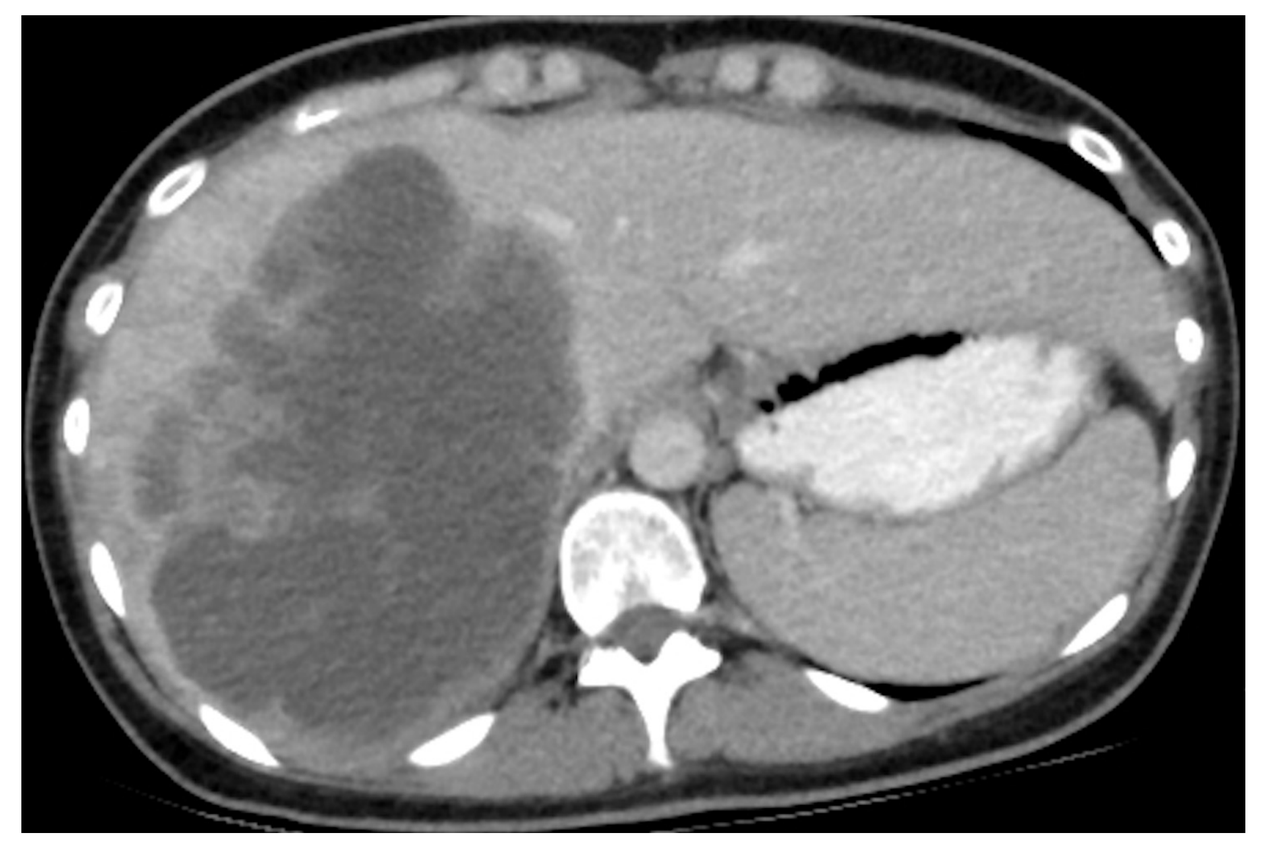

Figure 1. Liver metastasis 135 x 98 mm April 2019

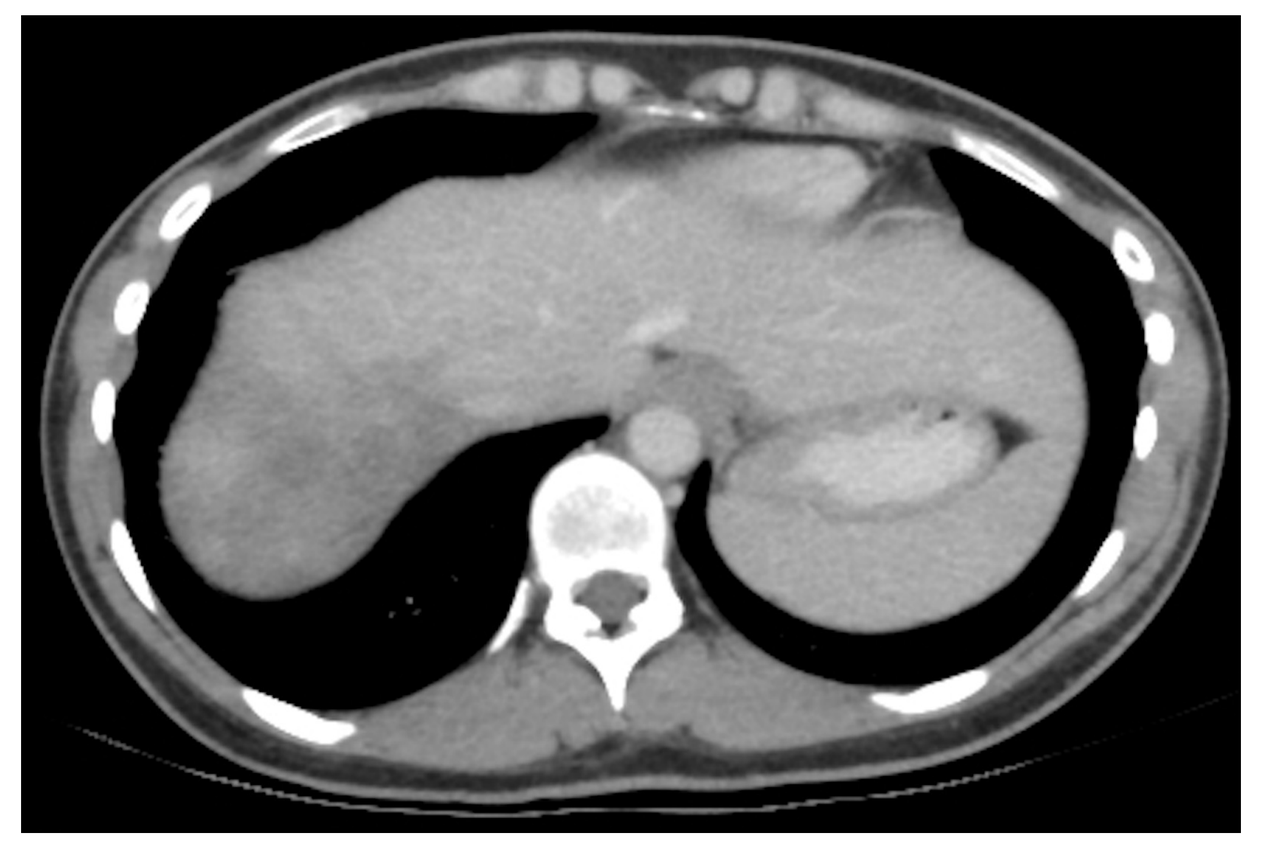

Figure 2. Liver metastasis 17 x 14 mm October 2019

Case 2

- Cell cycle pathway alterations occur in 49% of pancreatic ductal adenocarcinomas (PDAC) and may confer sensitivity to CDK4/6 inhibitors[3].
- A PDAC patient harbouring biallelic loss of *CDKN2A* treated on the palbociclib substudy, maintained stable disease in non-target lesions for >24 months.

Case 3, 4 and 5

- *BRCA* mutations are found in 3.6% of biliary tract tumours (*BRCA1* 0.6%, *BRCA2* 3%). PARP inhibitors induce synthetic lethality in BRCA mutated cells[4].
- Treatment on the olaparib + durvalumab substudy yielded two partial responses and one durable stable disease amongst two cholangiocarcinomas and one small bowel adenocarcinoma following detection of *BRCA2* alterations.

Case 6

- *Neuregulin 1 (NRG1)* gene fusion proteins are an important oncogenic driver, enriched amongst *KRAS* wild-type PDAC (8-10%)[5].
- *NRG1* is the predominant ligand of *ERBB3*. These fusion proteins drive tumour progression through aberrant *ERBB3* activation.
- Identification of an *ATP1B1-NRG1* gene fusion permitted compassionate access to seribantumab (an anti-HER3 monoclonal antibody); resulting in a partial response (Figure 3) and ongoing treatment for 8 months.

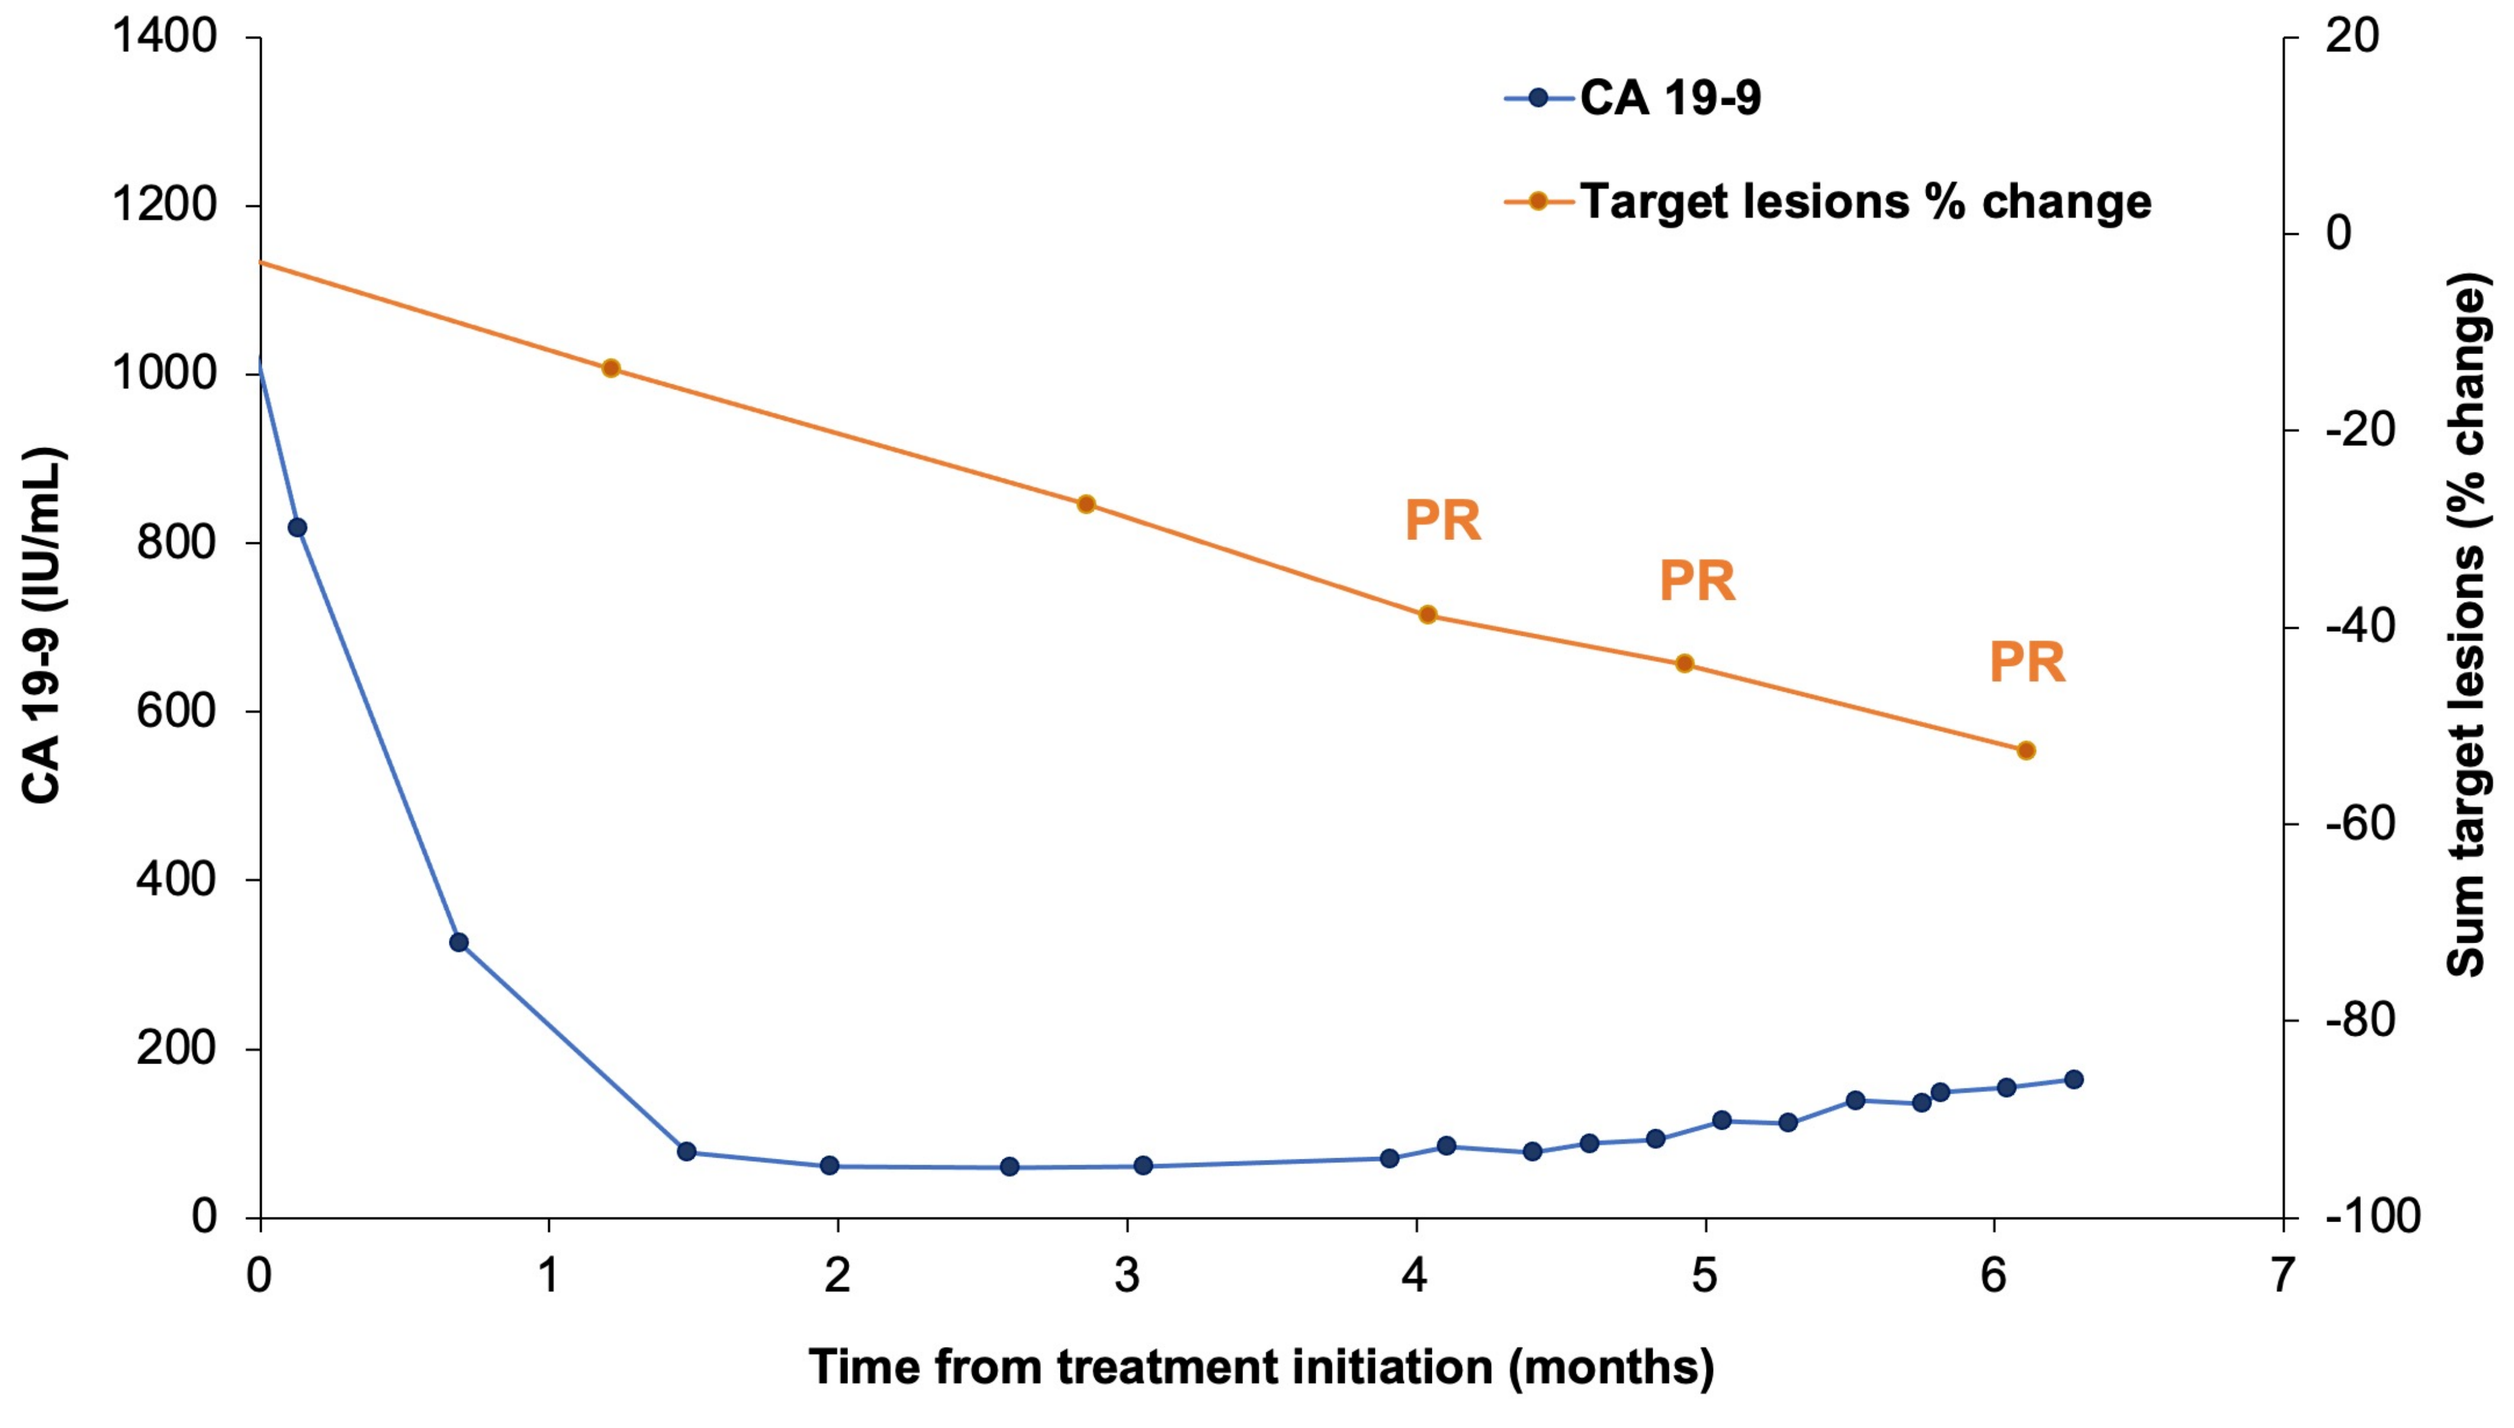
CA 19-9
Target lesions % change
PR
PR
PR
CA 19-9 (IU/mL)
Sum target lesions (% change)
Time from treatment initiation (months)
1400
1200
1000
800
600
400
200
0
20
0
-20
-40
-60
-80
-100
0
1
2
3
4
5
6
7

Figure 3. Seribantumab clinical activity in Case 6 (KRAS wt pancreatic adenocarcinoma with ATP1B1-NRG1 fusion)

Case 7

- *ERBB2* amplifications occur in 3-4% of mCRC and may identify patients for HER2-directed therapies[6].
- A patient with *ERBB2* amplified (25 copies) colorectal cancer was treated on the trastuzumab-emtansine substudy and maintained stable disease for 7 months.

Table 1. Summary of clinical benefit on biomarker-matched MoST substudy treatments

	Genomic Alteration	Tumour Type	Biomarker Matched Treatment	Best Overall Response	Treatment Duration	Duration of Response	Prior Lines of Treatment
1	*POLE P286R* mutation and high TMB (MSI-S)	Pancreatic acinar	Durvalumab + tremelimumab	Complete response	12 months	>24 months (ongoing)	2
2	*CDKN2A* loss	Pancreatic ductal	Palbociclib	Non-CR/Non-PD	26 months	26 months	4
3	*BRCA2* mutation	Cholangiocarcinoma	Olaparib + durvalumab	Partial response	31 months	28 months (ongoing)	1
4	*BRCA2* mutation	Cholangiocarcinoma	Olaparib + durvalumab	Partial response	9 months	7 months	2
5	*BRCA2* mutation	Small bowel adenocarcinoma	Olaparib + durvalumab	Stable disease	14 months	14 months	3
6	*ATP1B1-NRG1* fusion	Pancreatic, KRAS WT	Seribantumab	Partial response	9 months	3 months (ongoing)	3
7	*ERBB2* amplification	Colon, KRAS/BRAF WT	Trastuzumab-emtansine	Stable disease	6 months	6 months	2

Table 2. Potential actionable genomic alterations of GI patients in the MoST screening cohort[7]

	TOPOGRAPH highest actionability tier	No. of potential patients
Colorectal / Duodenal		
MSI high/MMR mutations	1B	10
ERBB2 overexpression	3	14
KRAS G12C	3	2
ATM	4	12
BRAF non V600	4	30
CHEK1	4	2
POLD1/POLE	4	1
ERBB3 overexpression	4	2
Gastric / GOJ / Oesophageal		
PTEN loss of function mutations	3	6
ERBB2 amplification*	3	8
EGFR/KRAS/MET/RICTOR amplification	4	25
Pancreas		
BRCA1/2	3	14
PALB2	3	3
BRAF V600E	4	1
CDKN2A	4	30
FANCG	4	1
NRG1 fusions	4	1
RNF43	4	6
STK11	4	7
Biliary / Gallbladder		
IDH1 R132	1B	5
FGFR2 fusions	2	11
ERBB2 amplification	3	0
BRAF V600E	3	0
ATM	4	0
Hepatocellular Carcinomas		
FGF19 amplification	4	0
Total GI cancer patients with actionble genomic findings		**191**

*only oesophageal cancers included as HER2 directed therapy is part of standard of care treatment

Conclusion

- CGP can identify rare, but therapeutically relevant genomic alterations with the potential to improve clinical outcomes for advanced, GI cancer patients.
- Future research should focus on how best to identify patients who will derive the greatest benefit from this precision oncology approach.

Acknowledgements

Thank you to the Garvan screening team, TKCC and CTC trials team. The MoST program was supported by Roche, Pfizer, Elevation Oncology and Astra Zeneca. Seribantumab was provided by Elevation Oncology through an Expanded Access Program.

References

1. Thavaneswaran S et al. Med J Aust. 2018 Aug; 209(8)
2. Guenther M et al. J Cancer Res Clin Oncol. 2018 Nov; 144(11)
3. Lin JC et al. J Clin Med. 2020 Dec; 9(12)
4. Spizzo G et al. ESMO Open. 2020 Jun; 5(3)
5. Aguirre AJ. Clin Cancer Res. 2019 Aug; 25(15)
6. Loree JM. JNCI. 2018 Dec; 110(12)
7. Frank PL et al. NPJ Precis Oncol. 2021 Jun; 5(1):58